PRELIMINARY REPORT OF CRENOLANIB IN THE TREATMENT OF ADVANCED PLATELET DERIVED GROWTH FACTOR A (PDGFRA) D842V MUTANT GASTROINTESTINAL STROMAL TUMOR (GIST)

M. von Mehren[1], M. Heinrich[2], E. Tetzlaff[1], K. Padavic-Shaller[1], M. Yu[1], C. Muralidhara[3], A.Ramachandran[3], H. Shah[3]
[1]Fox Chase Cancer Center, Philadelphia, PA; [2]Oregon Health and Science University, Portland, OR; [3]AROG Pharmaceuticals, Dallas, TX.

BACKGROUND

Many patients with advanced GIST treated with approved tyrosine kinase therapies have prolonged disease control with a median survival of 5 years. Rare subsets of GIST do not derive the same benefit from treatment. One such subset is GIST that carries a mutation in PDGFRA exon 18, D842V. In vitro, approved therapies do not cause a decrease in cell proliferation or loss of PDGFRA phosphorylation[1]. In clinical trials, available data suggests no response to standard therapies[2].

Crenolanib is a benzimidazole compound[3] being developed for the treatment of GIST patients with *PDGFRA*^{D842V}. Crenolanib is a potent and specific inhibitor of type III tyrosine kinases.

In CHO cell lines transiently transfected with *PDGFRA*^{D842V}, crenolanib inhibits the phosphorylation of the mutant PDGFRα with an IC_{50} of 9nM and IC_{90} of 44nM[4].

STUDY DESIGN

This is an open label phase II study conducted at 2 centers (FCCC and OHSU) (NCT01243346).

Key Inclusion criteria

- At least 18 years of age
- ECOG PS ≤ 1
- History of GIST with a documented PDGFRA D842V mutation
- Liver function tests $\leq$ 2X the ULN in the setting of liver metastases, and $\leq$ 1.5X the ULN with no liver metastases

Endpoints

- Primary endpoint: Response rate to crenolanib, measured by RECIST
- Secondary endpoints: 6-month PFS and evaluation of PK in this patient population with prior gastric resections.

Treatment Plan

- Crenolanib 200 mg po QD (4 weeks = one cycle)
- Dose reductions to 160mg QD and 100mg QD for toxicities
- PET at baseline and at 4 weeks recommended
- CT/MRI repeat imaging every 2 cycles

PATIENT CHARACTERISTICS

- To date, 7 patients (4 F, 3 M) have been accrued.
- All had metastatic disease in liver and/or mesentery/retroperitoneum.
- Best response to prior therapy was stable disease.
- Safety data is available in 6 patients and efficacy in 6 patients.

	Median (Range)
Age (years)	66 (46 – 76)
ECOG Performance status	
0	6
1	1
Years since diagnosis	5 (1 – 35)
Primary tumor site	
Stomach	3
Small bowel	
Mesentary	2
Unknown	1
Number of prior systemic therapies	3 (0 – 9)
Prior exposure to TKIs	
Imatinib	5 (71%)
Sunitinib	3 (43%)
Dasatinib	4 (57%)
Sorafenib	1 (14%)

TOLERABILITY

Dose reductions	2 pts
Total crenolanib therapy	19 – 125 days
Reasons for discontinuation	
Adverse events	0
Progressive disease	3

- Significant AEs included elevation of liver function tests and anemia.
- Anemia requiring transfusion was observed in 3 pts, and was ascribed to intratumoral bleeding, following no evidence of GI bleeding or hemolysis. These AEs have not been observed with crenolanib therapy in patients with other solid tumors including non-GIST sarcomas.
- Ascites (2 pts) and pleural effusions (1 pt) have also been observed, including hemorrhagic ascites in 1 pt.

	Adverse Events	Grade	Events (Patients)
Likely Related	Anemia	4	1 (1)
	GGT increased	1	1 (1)
	Nausea	1	1 (1)
	Diarrhea	1	2 (1)
Possibly Related	Anorexia	1	3 (3)
	Chills	1	2 (2)
	Nausea	1	2 (2)
	Diarrhea	1	4 (2)
		2	1 (1)
	Anemia	3	1 (1)
	AST increased	1	1 (1)
		2	1 (1)
	ALT increased	1	1 (1)
	GGT increased	3	1 (1)
	Alkaline Phosphatase Increased	1	1 (1)
		2	1 (1)
	Fever	1	1 (1)
	Peripheral Edema	1	1 (1)

PHARMACOKINETICS

- Serum pharmacokinetics samples were obtained pre dose and at 30 (± 10), 60 (± 15), 120 (± 15) minutes and at 4 (±1), 8 (±2), and 24 (±4) hours after crenolanib administration
- Analysis was performed by an isocratic high performance liquid chromatography assay with tandem mass spectrometry
- Crenolanib was rapidly absorbed, with a t_{max} of ~2 hours
- Serum trough concentrations of crenolanib (at 24hrs) were ~12% the peak concentration.

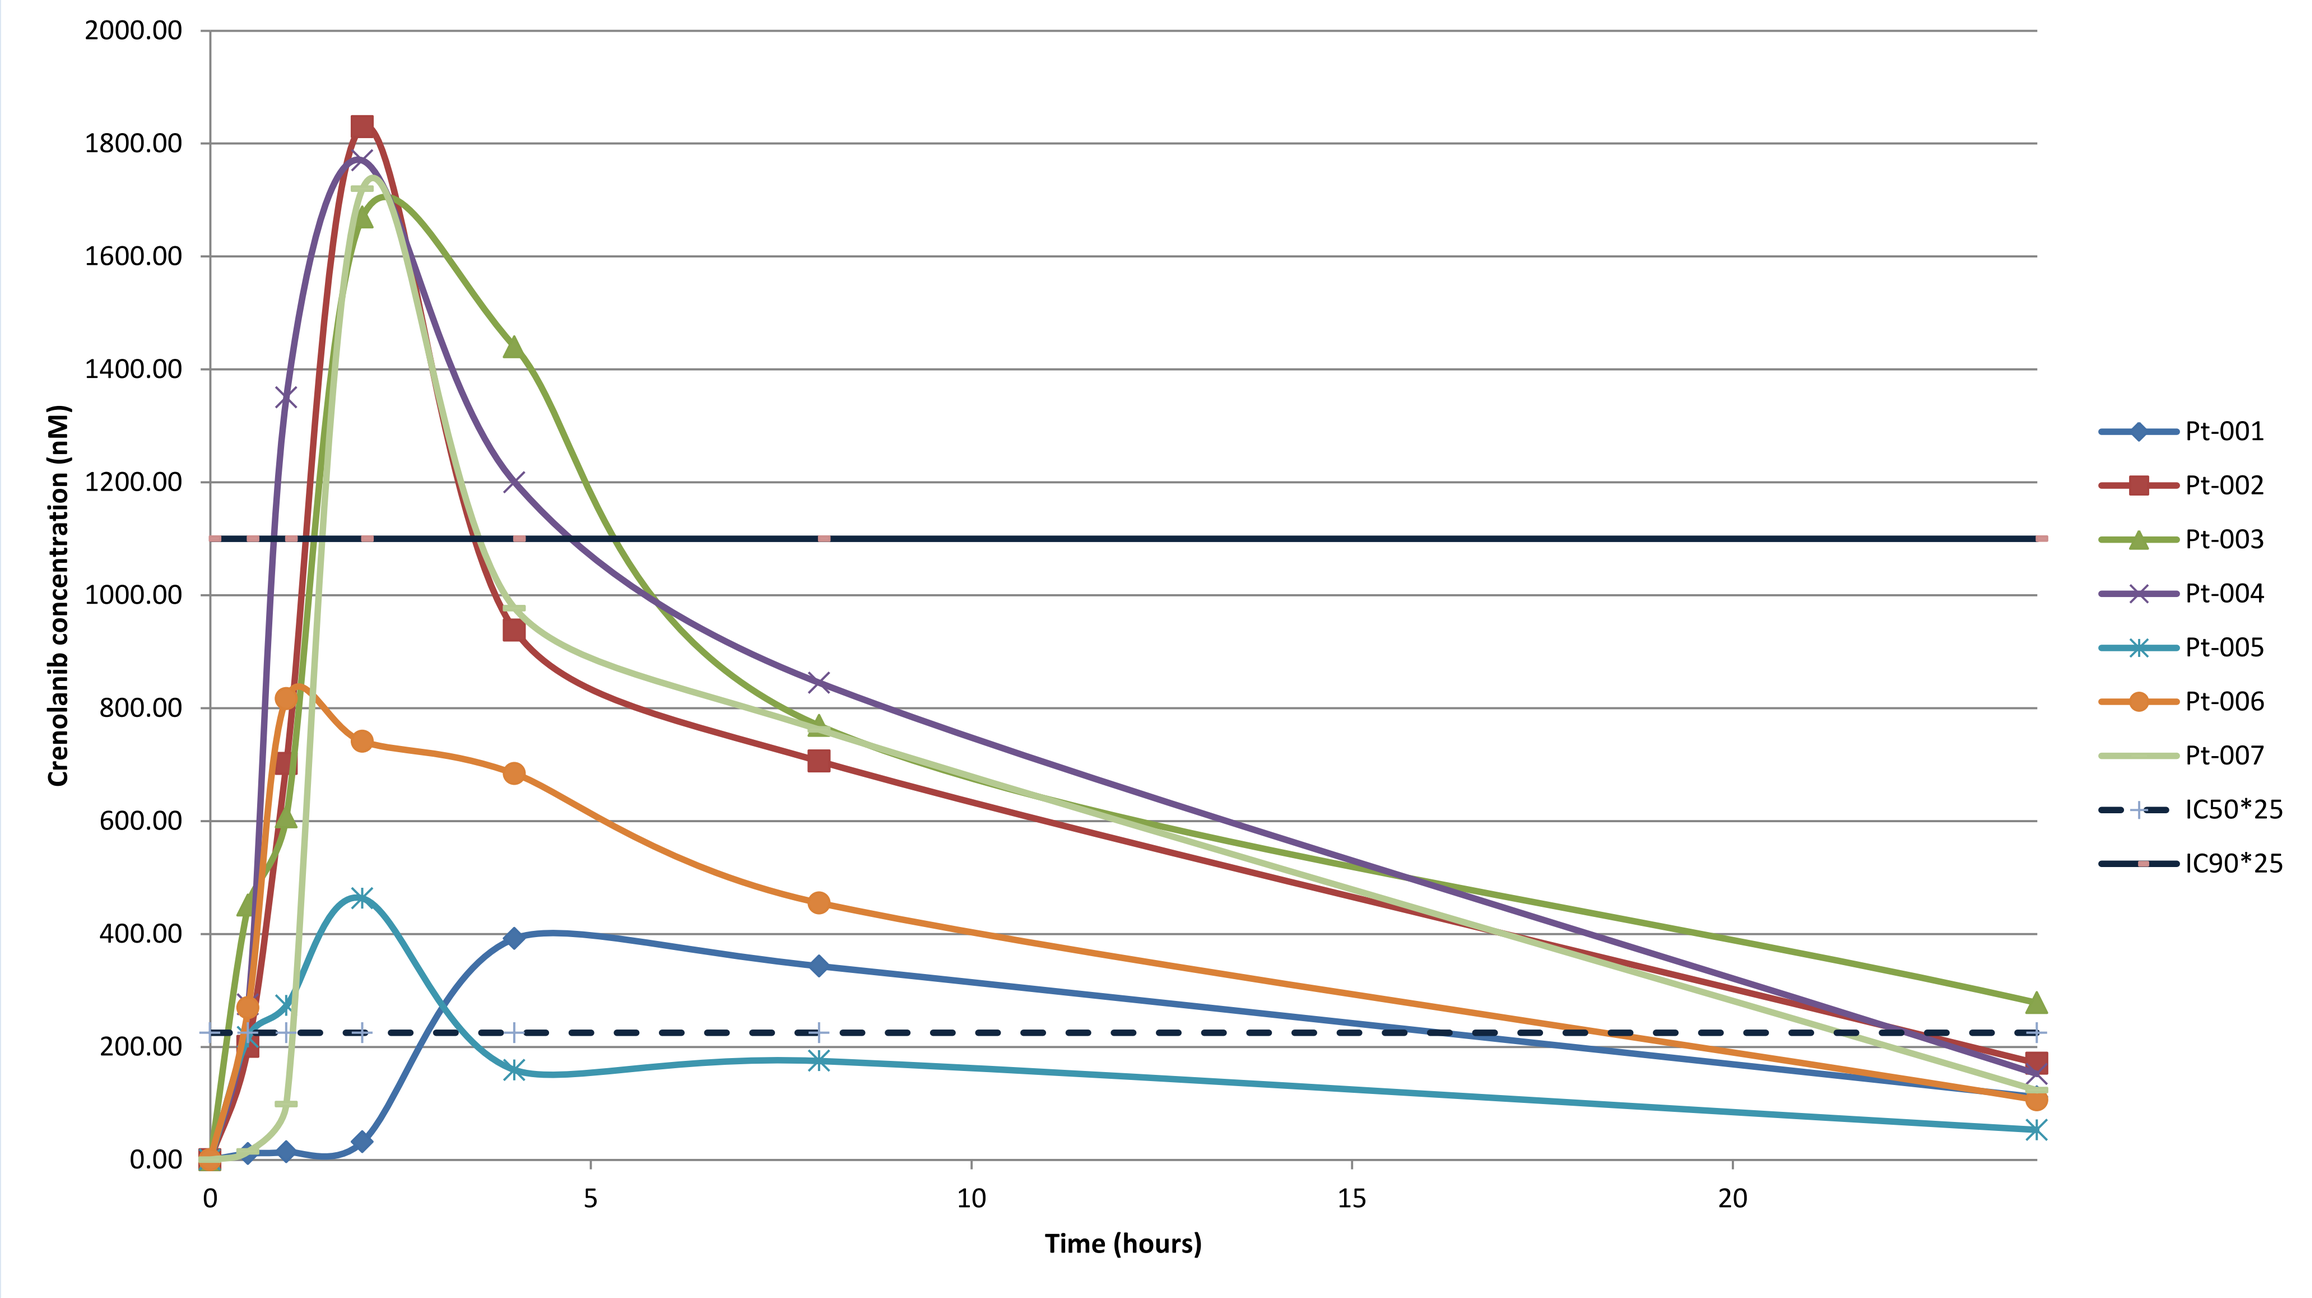
Crenolanib concentration (nM)
Time (hours)
Pt-001
Pt-002
Pt-003
Pt-004
Pt-005
Pt-006
Pt-007
IC50*25
IC90*25

- Serum concentrations of crenolanib achieved were around 4X the IC_{50} of crenolanib for PDGFRα, even at trough.
- Maximum inter-patient variability in C_{max} was ~4X.
- In the 4 patients where Day 15 crenolanib concentrations were available for assessment, only 1 patient showed significant accumulation at trough.

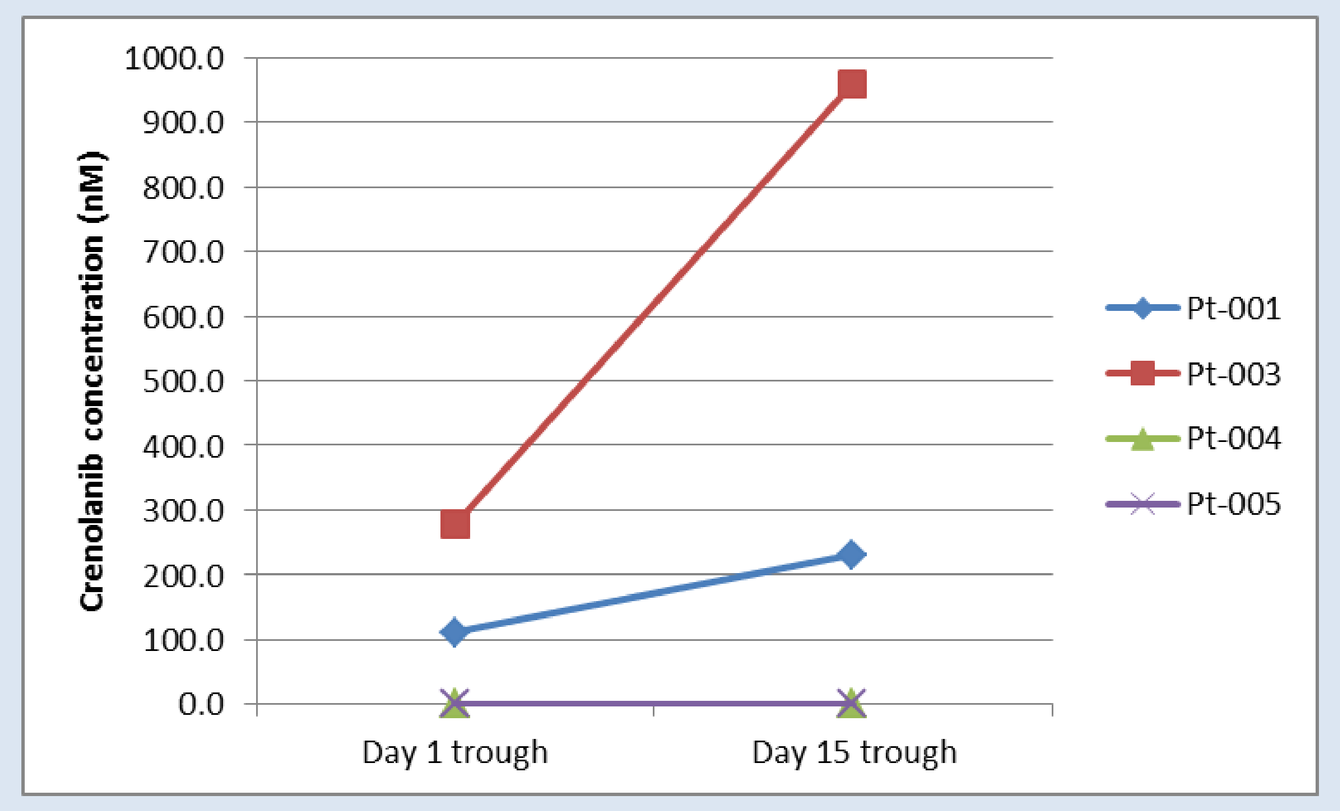
Crenolanib concentration (nM)
Day 1 trough
Day 15 trough
Pt-001
Pt-003
Pt-004
Pt-005

CRENOLANIB IS ABSORBED IN SPITE OF GASTRECTOMY

- Preclinical studies show that crenolanib is absorbed in the distal stomach[3].
- Four of seven patients treated had undergone prior gastrectomies.
- Absorption and PK characteristics in the gastrectomized patients were not significantly different from those in non-gastrectomized patients, suggesting that crenolanib may be absorbed in the small bowel in patients with gastric resection.
- Gastrectomy does not appear to impede the absorption of therapeutic concentration of crenolanib.

PARTIAL METABOLIC RESPONSE

Baseline

After 20 days of treatment with crenolanib, 200mg QD

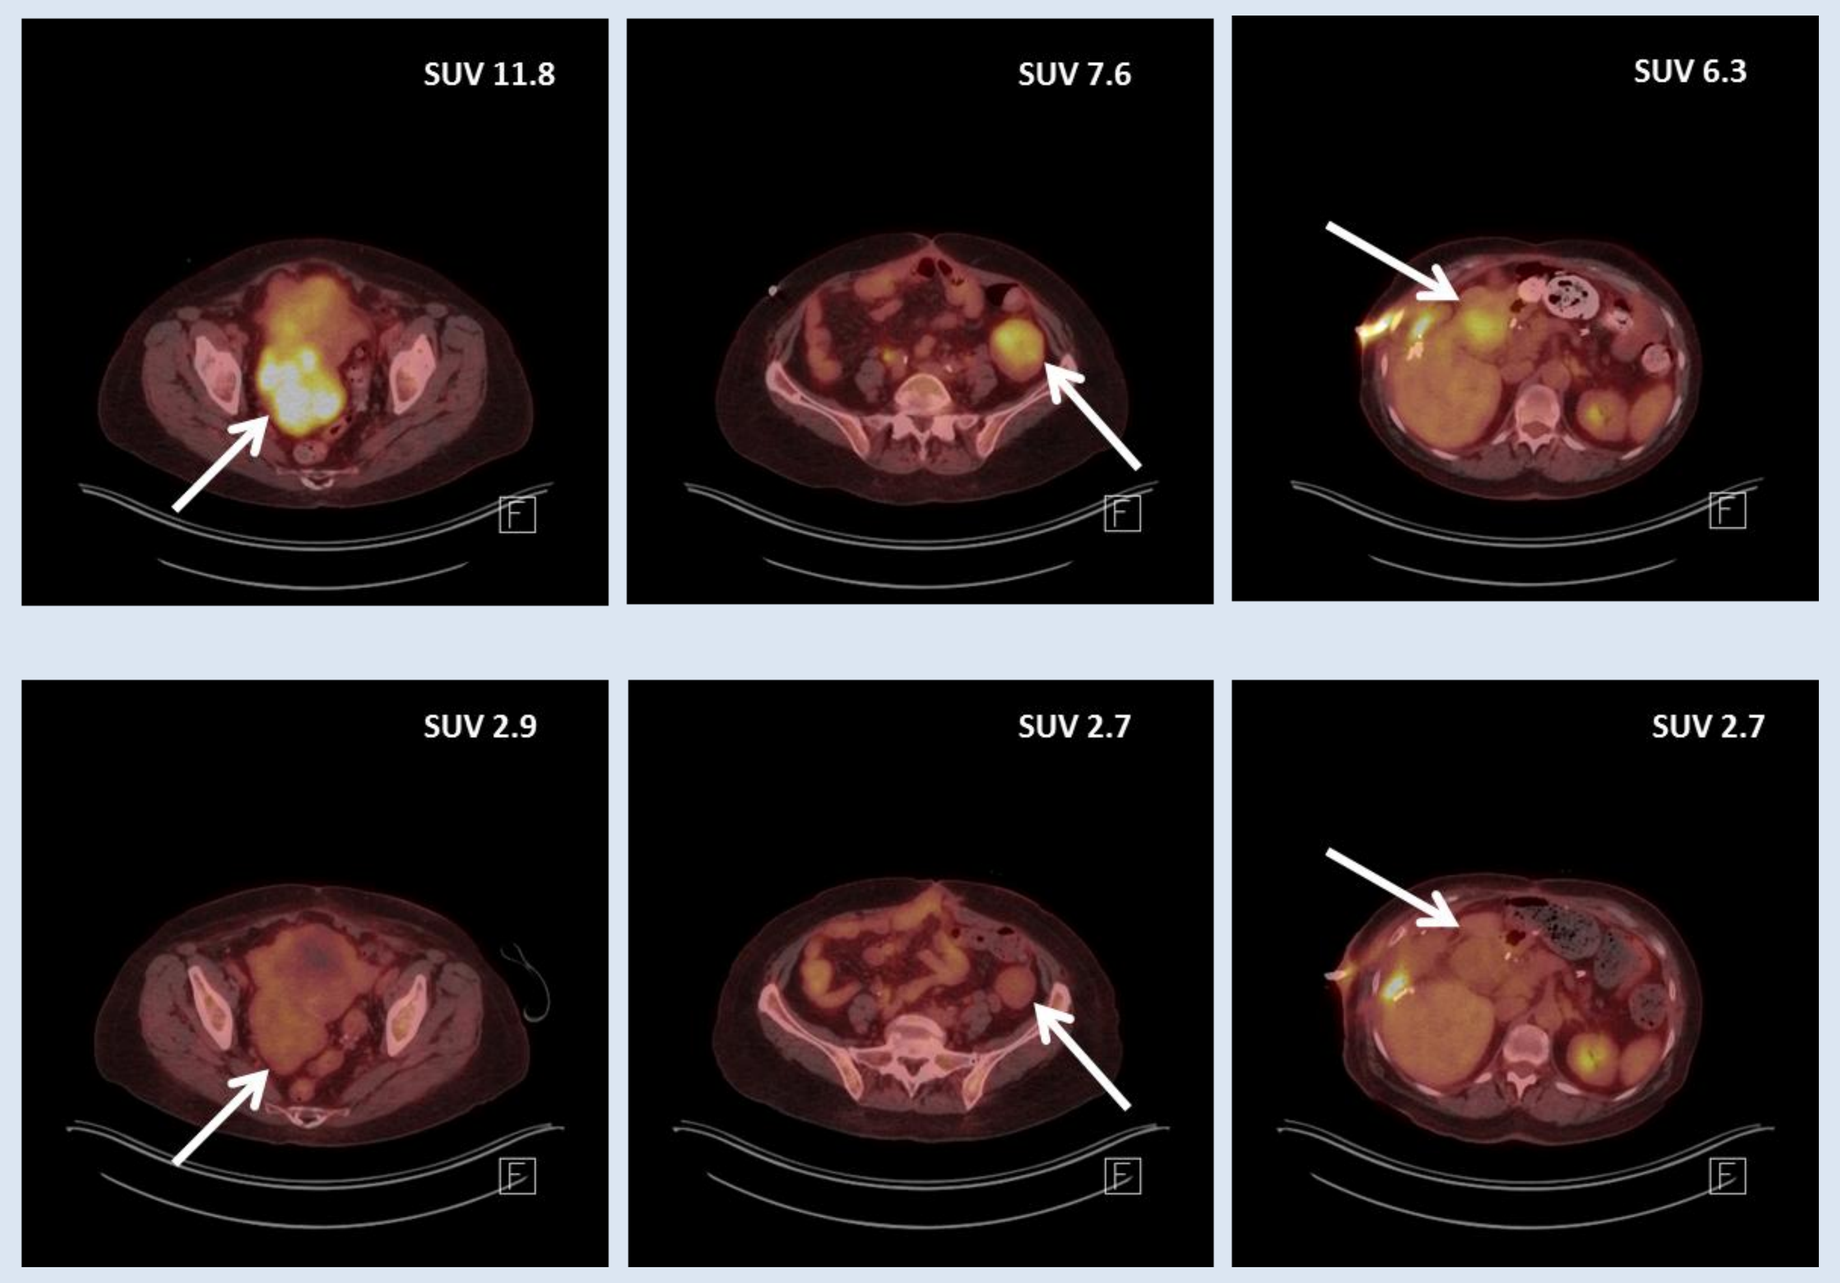
SUV 11.8
SUV 7.6
SUV 6.3
SUV 2.9
SUV 2.7
SUV 2.7

- 2 patients had metabolically active disease (SUV ≥ 10 in baseline PET scan) upon study entry.
- One patient (Pt-001) showed a marked reduction in glucose uptake (reduction in SUV from 11.8 to 2.8 in PET scan) after 28 days of entry into study (20 days of crenolanib exposure), with evidence of vascular collapse and necrosis in one lesion.
- No objective responses (CR or PR) have been observed to date.

CONCLUSIONS

- Crenolanib is the only available TKI with in vitro activity against *PDGFRA*^{D842V}.
- Absorption of crenolanib does not appear to be affected by gastrectomy.
- Toxicities have mirrored phase I experience , with nausea and vomiting managed with daily ondansetron administration.
- Two patients have experienced anemia attributed to bloody ascites, possibly related to crenolanib.
- Preliminary metabolic response was observed in one of seven patients treated.
- Accrual into this trial is ongoing.
- Future trials are planned to optimize the dose and schedule of crenolanib in this patient population.

REFERENCES

1. Dewaele, B., et al., Activity of Dasatinib, a Dual SRC/ABL Kinase Inhibitor, and IPI-504, a Heat Shock Protein 90 Inhibitor, against Gastrointestinal Stromal Tumor–Associated PDGFRAD842V Mutation. *Clin Cancer Res*, 2008. **14**(18): p. 5749-5758.
2. Biron, P., et al., Outcome of patients (pts) with PDGFRAD842V mutant gastrointestinal stromal tumor (GIST) treated with imatinib (IM) for advanced disease. *J Clin Oncol* (Meeting Abstracts), 2010. **28**(15_suppl): p. 10051.
3. Crenolanib Investigator's Brochure, *AROG Pharmaceuticals, LLC,* 2011.
4. Heinrich, M.C., et al., The effect of crenolanib (CP-868596) on phosphorylation of the imatinib-resistant D842V PDGFRA activating mutation associated with advanced gastrointestinal stromal tumors. *J Clin Oncol* (Meeting Abstracts), 2011. **29**(suppl;): p. abstr 10012.